External Assessment of Laboratories for the Measurement of Sirolimus by HPLC

Kirsty Denny[1], Denise A McKeown[1], Ron Stroshane[2], Atholl Johnston[1] and David W Holt[1].
Analytical Unit, St George's Hospital Medical School, London, UK[1] and Bioanalytical R&D, Wyeth Research, Collegeville, PA, USA[2].

INTRODUCTION

Therapeutic Drug Monitoring

Measurement of the immunosuppressive drug sirolimus (rapamycin, Rapamune®) is a regulatory requirement in Europe and is recommended in several clinical settings in the USA (e.g. hepatic impairment; when strong inducers or inhibitors of CYP3A4 are introduced or discontinued; when cyclosporin is markedly changed or discontinued).

Proficiency Testing (PT)

Previously, we have highlighted the need to monitor and document laboratory proficiency both to compare results produced by different analytical techniques and to ensure consistency of results between multiple testing sites. We have already devised a successful challenge to assess laboratory competence for the measurement of sirolimus using an investigational immunoassay based on microparticle enzyme immunoassay technology (Abbott, IL, USA).[1] This study extrapolates this experience to produce a PT study to assess the ability of laboratories to measure sirolimus by high-performance liquid chromatography (HPLC), using either ultraviolet (UV) or mass-spectrometric (MS) detection. Results have previously been presented from this study when only 49 laboratories were involved.[2]

AIM

For the last 4 years HPLC was the only analytical technique available to measure sirolimus. We report a study to assess the ability of over 150 laboratories located world wide (Figure 1) to measure sirolimus by HPLC, using either UV or MS detection. The centres were supporting target concentration dosing in multicentre clinical studies of sirolimus.

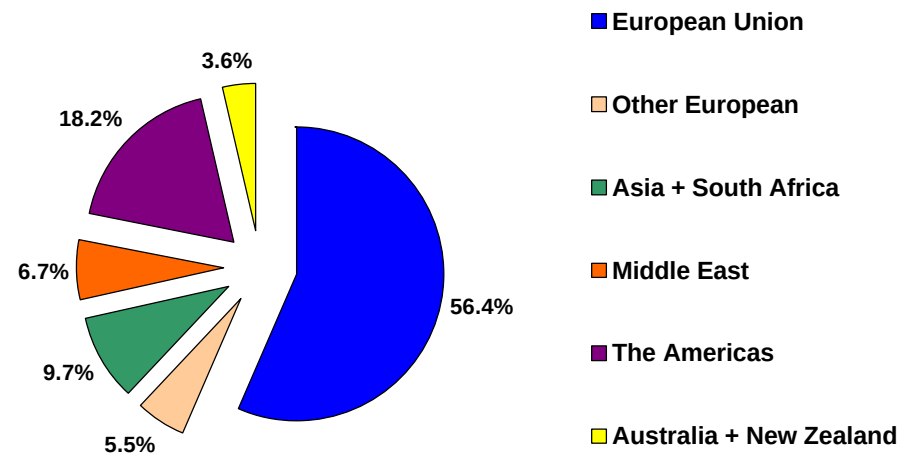
3.6%
18.2%
6.7%
9.7%
5.5%
56.4%
European Union
Other European
Asia + South Africa
Middle East
The Americas
Australia + New Zealand

Figure 1: Global Distribution of Study Centres

REFERENCES

1. Jones K, Johnston A, Holt DW. *Clin Ther* 2000:22 (Suppl B):B122-132.
2. Denny K, Lee TD, Johnston A, Hick D, Holt DW. Ther Drug Monit 2001: 23:465
3. Holt DW, Lee T, Jones K, Johnston A. *Clin Chem* 2000:46 (8):1179-1183.

METHODS

The laboratories were required to measure sirolimus in a set of 78 blinded EDTA-anticoagulated whole blood samples, packaged as 5 batches. Each set comprised aliquots of 4 spiked samples 7, 15, 25 (L, M, H) and 200ng/mL (out of range, OOR), 3 blood pools from patients receiving the drug as part of their immunosuppressive therapy following transplantation (P1 to P3), a set of calibrators spanning the range 0 to 50ng/mL (C1 to C5) and a sirolimus-free blood sample (Blank). The sirolimus concentrations of these samples was defined by measuring multiple replicates (L, M, H, OOR and pools n=10; C1 to C5 and Blank n=5) by a validated HPLC/MS/MS assay.[3] Centres analysed each batch on separate days, diluting the OOR samples to obtain an exact concentration. The samples were designed to assess calibration accuracy, precision (within & between batch), specificity and calibration curve linearity (Figure 2 & 3).

Performance was acceptable if:

- Imprecision (coefficient of variation, CV) and inaccuracy
 < 25% for sirolimus concentrations = 7.5ng/mL
 < 20% for sirolimus concentrations > 7.5ng/mL
- Blank identified and reported as below assay LLOQ
- OOR identified and diluted to obtain a result within 20% of the defined concentration
- At least 2/3 of results passed inaccuracy criteria (49/73 samples excluding blanks)

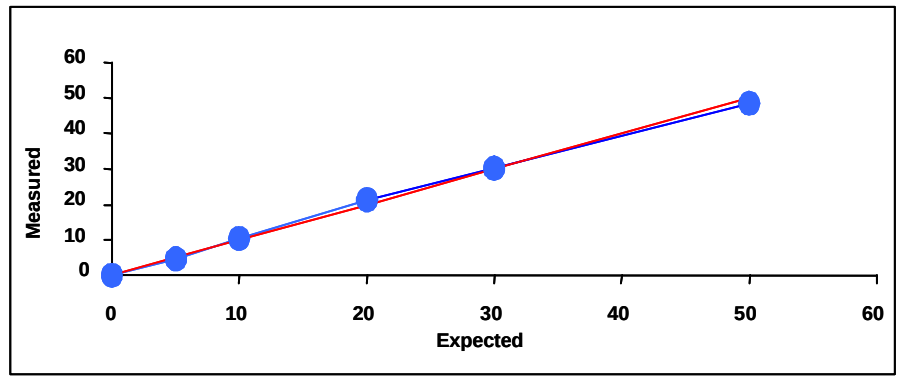
Measured
Expected
0
10
20
30
40
50
60

Figure 2: Calibration curve for a centre using UV detection demonstrating good linearity. Centre results (blue line) and defined values (red line).

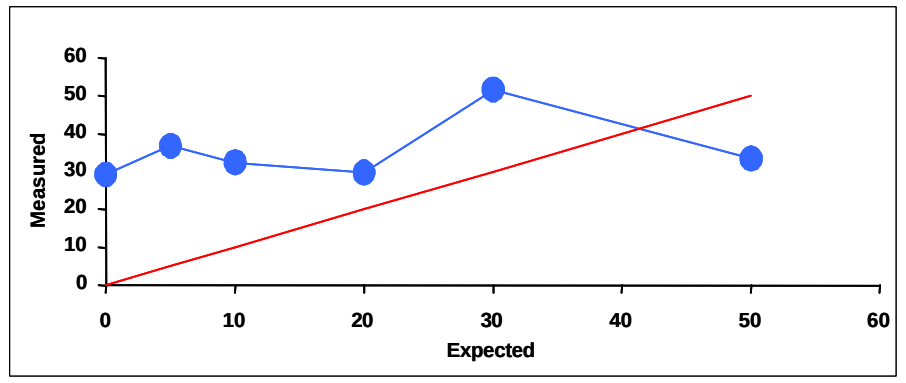
Measured
Expected
0
10
20
30
40
50
60

Figure 3: Calibration curve for a centre using UV detection demonstrating poor linearity. Centre results (blue line) and defined values (red line).

RESULTS

Many laboratories have been successful in analysing the 78 samples; 106 laboratories (64 UV, 42 MS) passed and a further 18 centres were approved after subsequent assessments.

Result summary for the 106 successful centres:

- The mean within and between assay imprecision (%CV), for the spiked (L, M, H) and pooled patient samples was = 9% (median < 8%) (Figure 4)
- For the calibrators, spiked samples (L, M, H, OOR) and the pooled patient samples, the mean inaccuracy was < 9% (median < 3%) (Figure 5)
- The mean number of results > 20% from the defined value (n error) was 9 (median = 9)

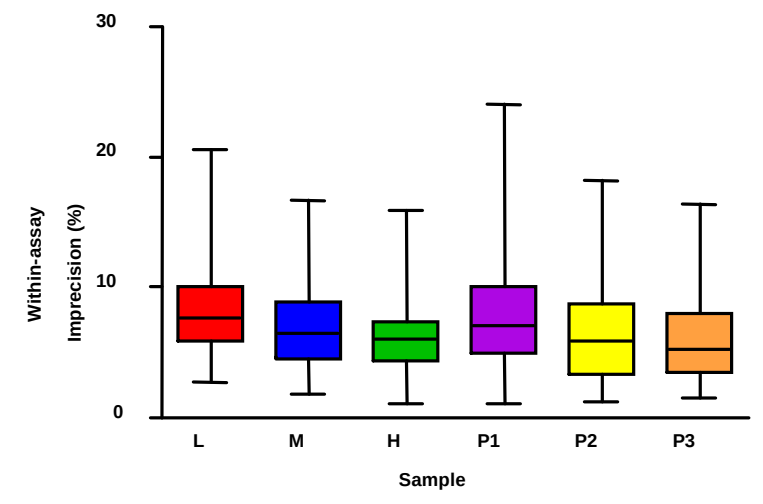
Within-assay
Imprecision (%)
0
10
20
30
L
M
H
P1
P2
P3
Sample

Figure 4: Within-assay imprecision for the L, M, H spiked samples and the 3 pooled patient samples (concentration range 5 to 30ng/mL).

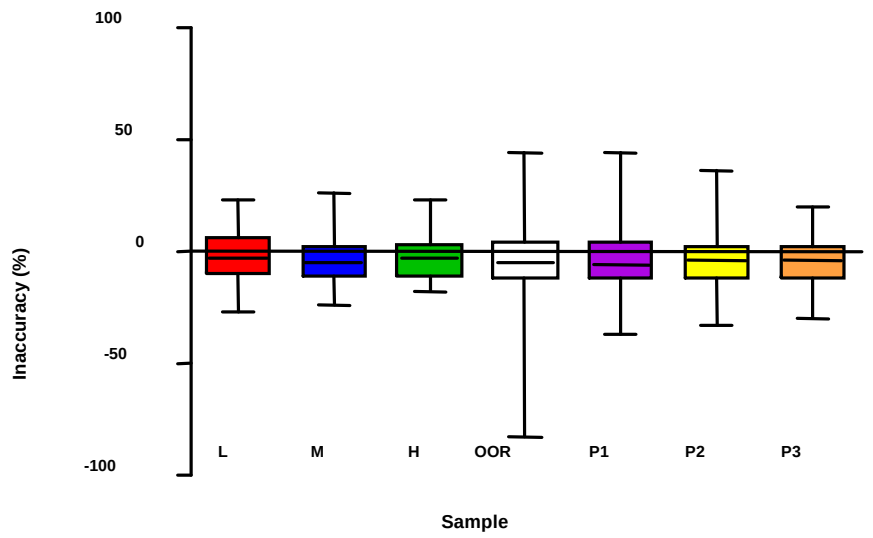
Inaccuracy (%)
100
50
0
-50
-100
L
M
H
OOR
P1
P2
P3
Sample

Figure 5: Inaccuracy for the L, M, H, OOR spiked samples and the 3 pooled patient samples (concentration range 5 to 200ng/mL).

CONCLUSION

This pre-study PT challenge has demonstrated a method, implemented internationally, to assess assay performance for the measurement of sirolimus by HPLC. Data from this study have been used to ensure consistency of results from multicentre clinical studies.